MSc. Martine H.P. Crins [a], dr. Caroline Terwee [b], prof. dr. Rene Westhovens [c,d], dr. Dirkjan van Schaardenburg [e], dr. N. Smits [f], MSc. Johan Joly [c,d], prof. dr. Patrick Verschueren [c,d], MSc. Kristien van der Elst [c,d], prof. dr. David Cella [g], dr. Karon F. Cook [g], prof. dr. Joost Dekker [h], prof. dr. Maarten Boers [b,i], dr. Leo D. Roorda [a]

[a] Amsterdam Rehabilitation Research Center | Reade, Amsterdam, The Netherlands, [b] Department of Epidemiology and Biostatistics, VU University Medical Center, Amsterdam, The Netherlands, [c] Department of Development and Regeneration, KU Leuven, Leuven, Belgium, [d] Rheumatology, University Hospitals Leuven, Leuven, Belgium, [e] Jan van Breemen Research Institute | Reade, Amsterdam, The Netherlands, [f] Department of Clinical Psychology and Department of Methodology, VU University Medical Center, Amsterdam, The Netherlands, [g] Department of Medical Social Sciences, Northwestern University Feinberg School of Medicine, Chicago, IL, USA, [h] Department of Rehabilitation Medicine and Department of Psychiatry, VU University Medical Center, Amsterdam, The Netherlands, [i] Amsterdam Rheumatology & Immunology Center, VU University Medical Center, Amsterdam, NL.
Correspondence: m.crins@reade.nl

Validation of the Dutch-Flemish PROMIS Pain Behavior and Pain Interference Item Banks in Patients with Rheumatoid Arthritis

BACKGROUND

In the assessment of patients with rheumatoid arthritis (RA) it is important to measure pain behavior and pain interference.

AIM

The aims of current study were to calibrate the Dutch-Flemish translation of the PROMIS Pain Behavior (DF-PROMIS-PB) and Pain Interference (DF-PROMIS-PI) item banks in Dutch and Flemish RA patients, and to evaluate the cross-cultural validity, reliability, and construct validity.

METHODS

- 1181 Dutch and 650 Flemish RA patients, 68% female, mean (SD) age: 56 (13) y
- Web-based or paper-and-pencil survey (each 50%) including full DF-PROMIS-PB and DF-PROMIS-PI item banks
- DF-PROMIS-PB: 39 items, 6-point Likert scale
- DF-PROMIS-PI: 40 items, 5-point Likert scale
- Calibration: Item Response Theory (IRT) modeling; confirmatory factor analysis to evaluate unidimensionality; fit of a graded item response model (GRM)
- Differential Item Functioning (DIF) for gender, age (median split), administration mode (paper vs. digital), language (Dutch vs. Flemish) and diagnoses (RA vs. chronic pain (a sample of 1140 patients from a previous study))
- By ordinal regression models with a McFadden's R^2 change of 2% as critical value

RESULTS

- DF-PROMIS-PB and DF-PROMIS-PI demonstrated good fit to a one-dimensional model:
 - CFI = 0.976; 0.997 resp. and TLI = 0.975; 0.997 resp.
 - First factor: 50% (DF-PROMIS-PB) and 80% (DF-PROMIS-PI) of the total questionnaire variance
- 14 of 741 (1.9%) DF-PROMIS-PB item pairs and 12 of 780 (1.5%) DF-PROMIS-PI item pairs marked as possibly locally dependent
- Threshold-parameters range from -1.7 to 3.8 for DF-PROMIS-PB and from -0.5 to 5.4 for DF-PROMIS-PI (see the Item Information Curves (IIC) in figure 1)

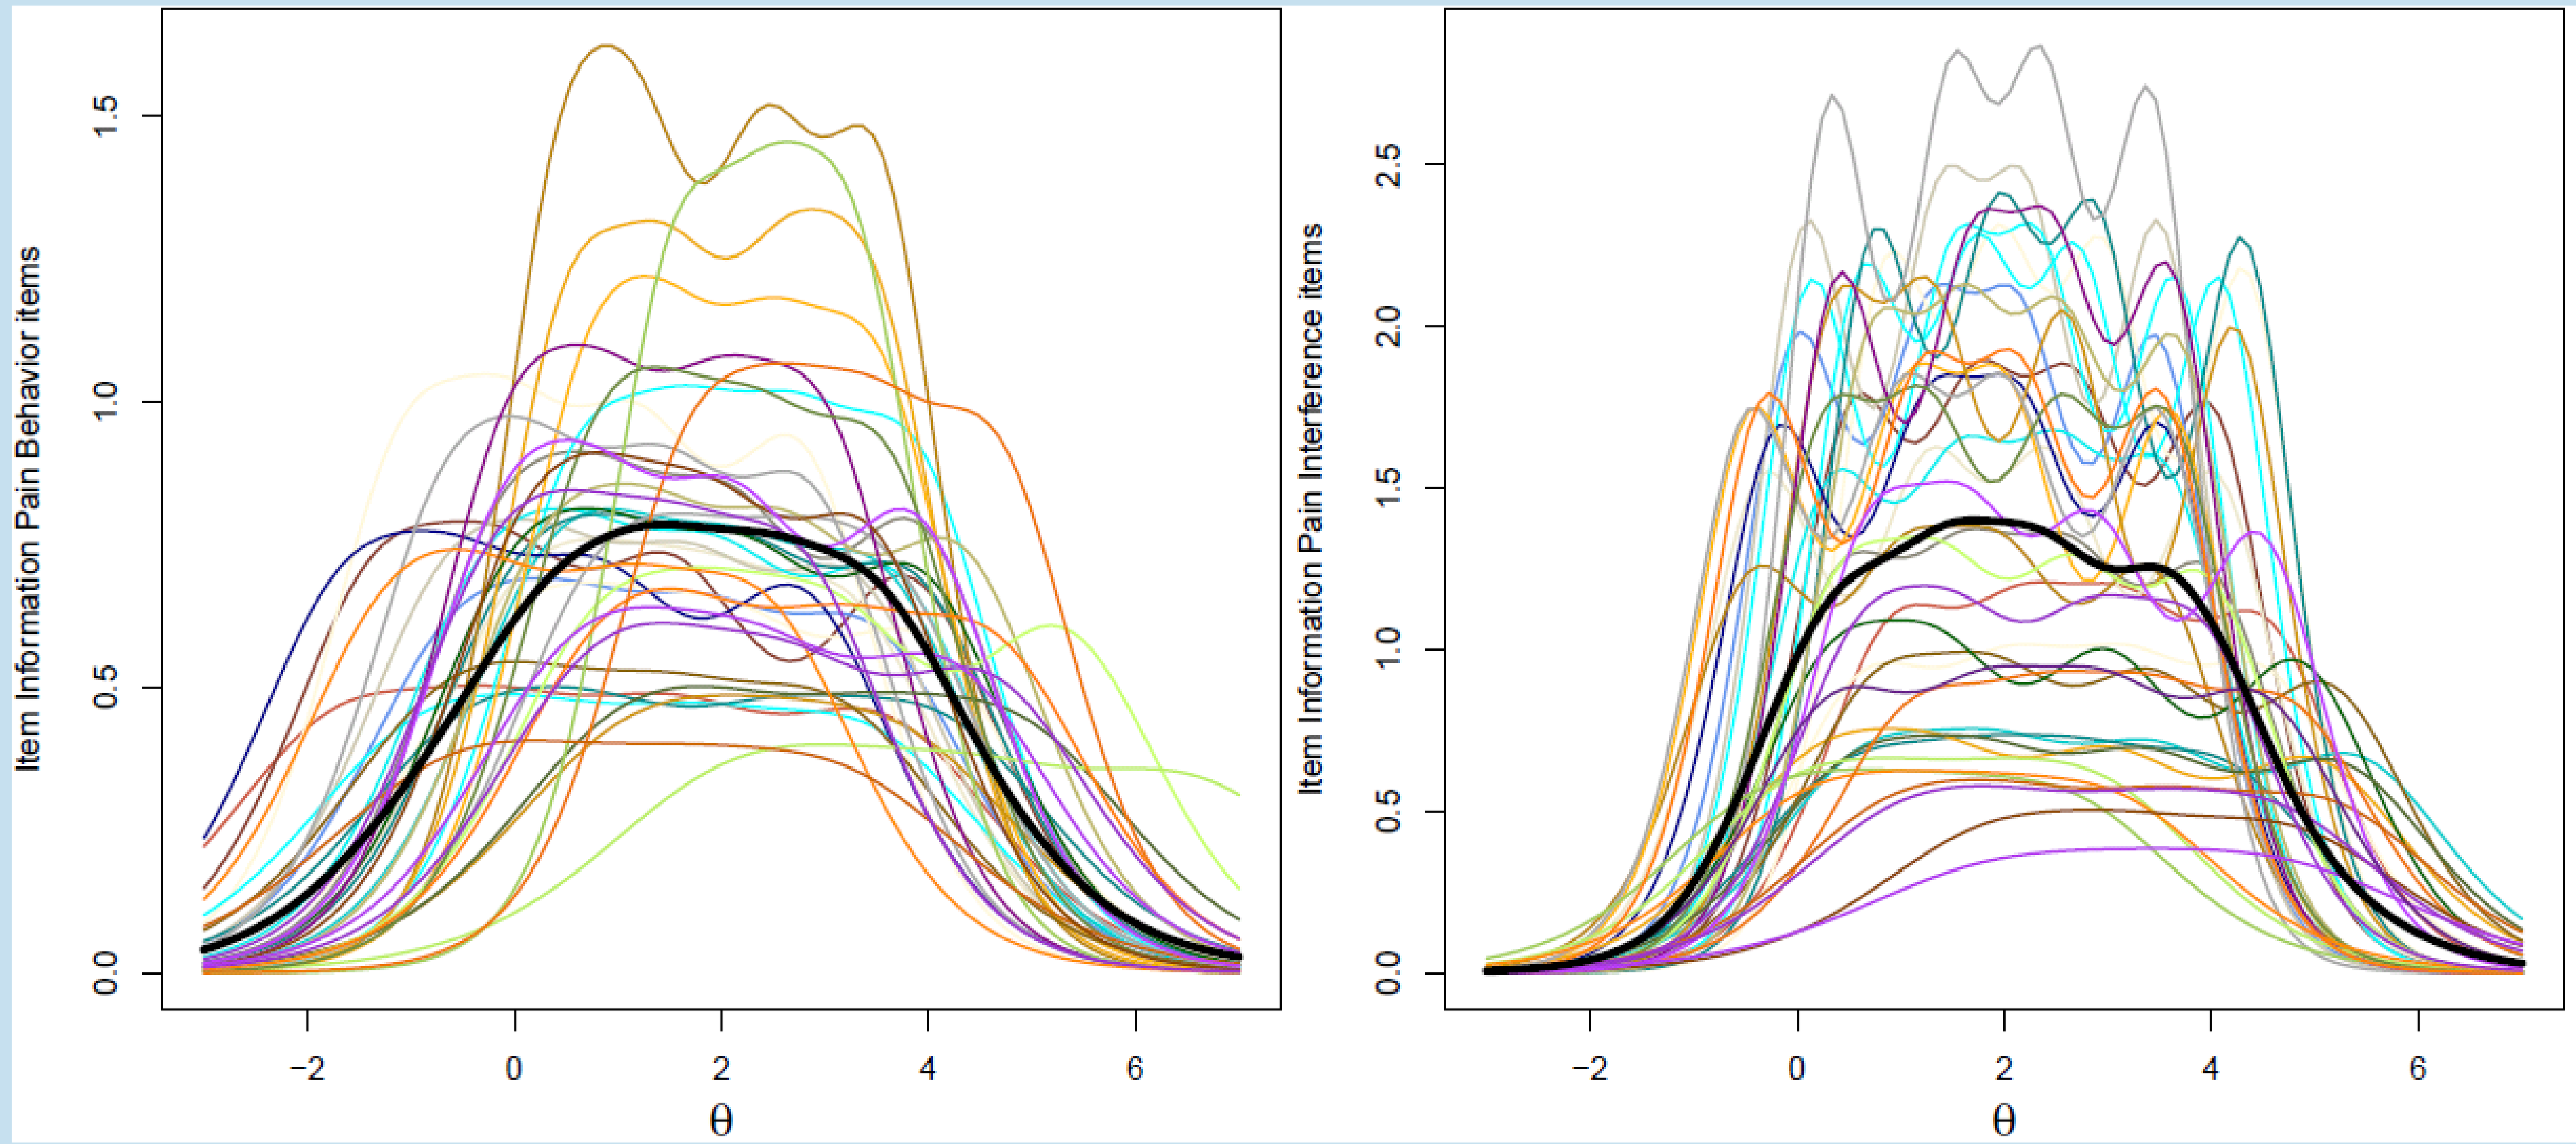
Item Information Pain Behavior items
θ
Item Information Pain Interference items
θ

***Figure 1**: Left: IIC of DF-PROMIS-PB. Right: IIC of DF-PROMIS-PI. The thick line is the average item information*

RESULTS

- Cronbach's alpha=0.985 (DF-PROMIS-PB); 0.989 (DF-PROMIS-PI)
- No DIF with respect to age, gender, and administration mode
- No DIF with respect to diagnosis
- 1 DIF item with respect to language:
 - DF-PROMIS-PB DIF item: PAINBE16 (When I was in pain I appeared upset or sad / Wanneer ik pijn had, zag ik er overstuur of verdrietig uit)

CONCLUSIONS

- **DF-PROMIS-PB and the DF-PROMIS-PI fit a GRM**
- **Nearly all Dutch item parameters match the US item parameters and likely Dutch-specific item calibrations are not needed**
- **DF-PROMIS-PB and DF-PROMIS-PI can be used to develop two Computer Adaptive Tests (CATs) for measuring pain behavior and pain interference respectively, in Dutch and Flemish RA patients**

www.dutchflemishpromis.nl